ECONOMIES OF SCOPE AND SCALE IN PUBLICLY FUNDED BIOMEDICAL AND HEALTH RESEARCH

Sue Guthrie[1], Karla Hernandez-Villafuerte[2,3], Sarah Parks[1], Alexandra Pollitt[1], Enora Robin[1], Jon Sussex[1], Steven Wooding[1]

[1] RAND Europe, [2]Office of Health Economics, [3]Current affiliation - German Cancer Research Center (DKFZ)

K.HernandezVillafuerte@dkfz.de/ Tel: +49 (0)6221421913

1. BACKGROUND

- Publicly funded biomedical and health research is expected to achieve the best return possible for taxpayers and for society generally
- Economies of scale/scope: More and/or higher quality outputs if undertaken in centres with a large number of researchers and/or a variety of research activities

Is biomedical and health research more productive if concentrated or if dispersed?

2. METHODS

- Literature reviews
 - ✓ Economies of scale and scope in medical research
- 23 interviews with UK MRC funded researchers
 - ✓ Basic/clinical, early/late career, large/smaller institutions
- Two case studies: (1) Centre for Regenerative Medicine, Edinburgh (2) MRC Institute of Metabolic Science, Cambridge
- Economic model identified: Econometric analysis
 - ✓ Bibliometric data: the number of articles, journal, institution of affiliation and co-authors
 - ✓ Survey to 500 researchers: size of the research group
 - ✓ Method: Segmented linear regression model

3. RESULTS - Literature Review

Table 1. Numbers of papers finding (dis-)economies of scale in biomedical and health research – by type of research group whose productivity is being analysed

	PI*	RT**	DT ***	University Research Institute	Other	Total
Diminishing returns to scale / Diseconomies of scale	2	3	1	3	-	9
Increasing returns to scale / Economies of scale	1	3	2	9	-	15
Constant returns to scale	1	3	1	-	2	7
Inverse U-shape	-	2	-	1	-	3
Not clear	2	1	3	-	2	8
Sub-total	**6**	**12**	**7**	**13**	**4**	**42**
Articles that suggest both diseconomies and economies of scale						
-Multi-product cost functions applied	-	-	-	4	-	4
-Other methodology applied	-	1	-	4	-	5
Total	**6**	**13**	**7**	**21**	**4**	**51**

*PI = Principal Investigator. **RT = Research Team = Sub-departmental laboratory/ research unit. ***DT = University department or research centre inside a bigger organization. A department is typically composed of a number of research teams.

4.RESULTS - Interviews

- Researchers distinguish between different levels of co-location (group, institute, town)
- Ideal group size 5-20 researchers.
- Ideal institute size 20-40 groups (with outliers)
- Many different perspectives on collaboration

Figure 1. Principal communications tools used by the researchers: (1) face-to-face meetings and (2) emails

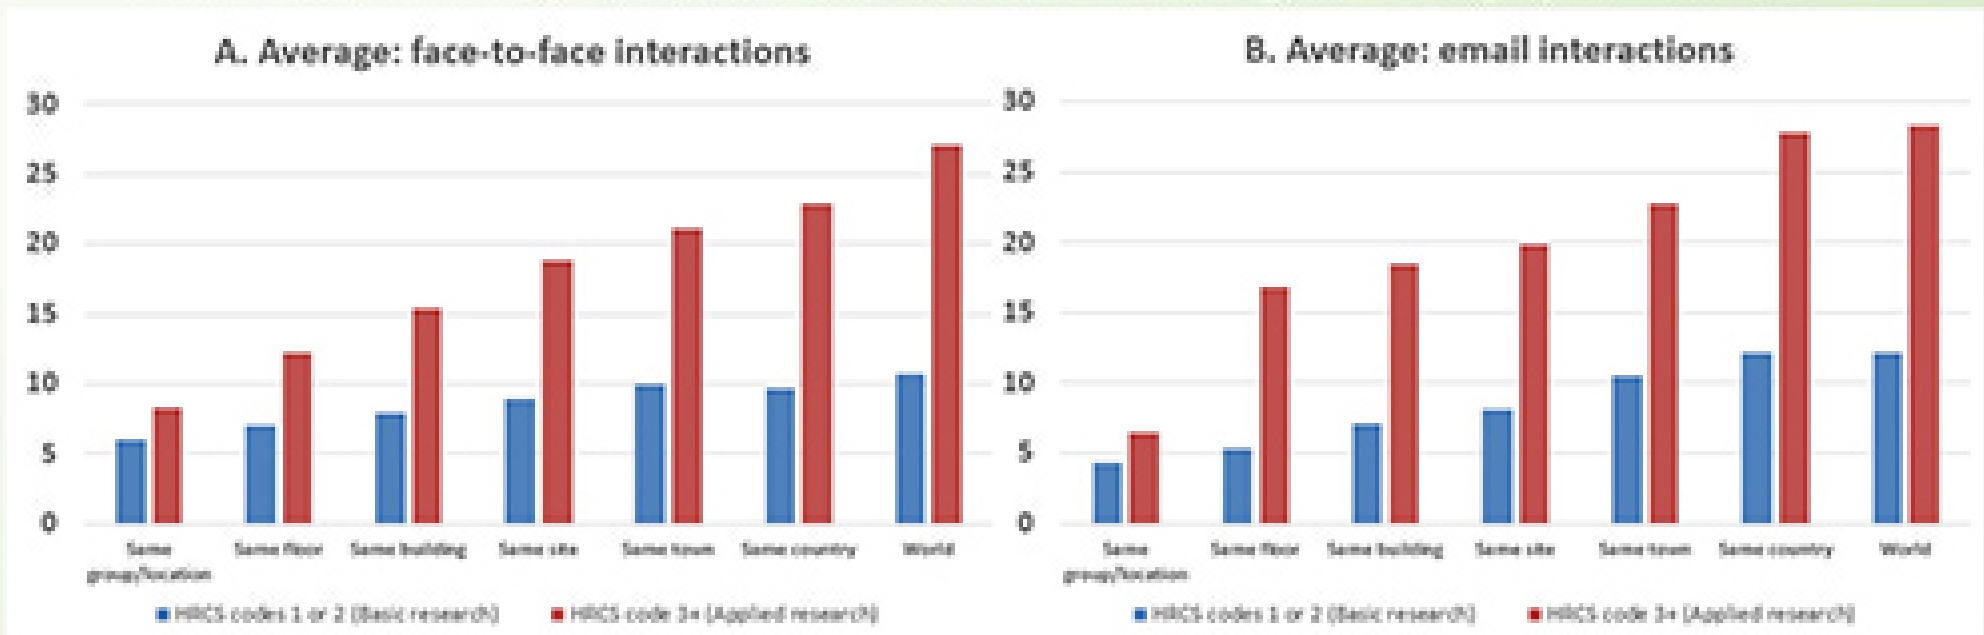
A. Average: face-to-face interactions
B. Average: email interactions
Same group/location
Same floor
Same building
Same site
Same town
Same country
World
HRCS codes 1 or 2 (Basic research)
HRCS code 3+ (Applied research)

5.RESULTS – Case studies

✓	×
• Shared building/facilities force interaction • Shared vision/branding has helped • Role of students – e.g. joint supervisors, informal discussions	• Bringing together diverse researchers can isolate them from their peers • Structure of university and research system can be barriers • Challenges of expansion

6. RESULTS – Econometric Analysis

Figure 2. Segmented linear regression model (Without outliers)

Intercept		2.640	***
Scaleproxy: Before saturation point		0.099	***
Scaleproxy: After saturation point		-0.008	NA
Scopeproxy: Research Activity Codes	2	0.118	
	3	0.227	•
	more than 3	0.513	***
Coauthors		0.001	***
Biological Age	40-49	0.046	
	50-59	0.022	
	60 or more	0.373	•
Academic Age		-0.001	
Sex	Fem.	-0.103	
Saturation point value		**12.73**	***

Dependent Variable

Total of Articles adjusted by quality and number of coauthors:

$$Y_j^2 = \sum_{i=1}^{n_j} \frac{Quality(MNCS)_{ij}}{authors_{ij}}$$

Where $Quality_{ij}$ = mean normalized citation score (MNCS) of paper *i* ($i = 1,\ldots,n_j$) in which the PI *j* is listed as co-author. Authors = number of co-authors in the paper *i* ($i = 1,\ldots,n_j$) in which PI *j* is listed.

Independent Variables

ScaleProxy: variable used in the analysis was defined as the sum of all PI's team members (PhDs students, Postdocs and others).

Co-authors: Total number of unique names that during the period of the bibliometric analysis are listed as co-authors of the PI's articles. This reflects the network of the PI's collaborations.

Biological age: (1) less than 40, (2) 40-49 (3) 50-59, and (4) 60 and more

Academic age – Experience : Number of Years between PhD graduation and 2016. This aim as reflect years of experience.

$$Scope\ Proxy_{dimension} = \begin{cases} \text{If the PI has all grants assigned to one code} & \rightarrow 1 \\ \text{If the PI has grants assigned to two different codes} & \rightarrow 2 \\ \text{If the PI has grants assigned to three different codes} & \rightarrow 3 \\ \text{If the PI has grants assigned to al least four different codes} & \rightarrow 4 \end{cases}$$

7. CONCLUSION

Economies of Scale: The empirical literature is varied both in method and findings. *Taken as a whole*: Greater likelihood of finding positive economies of scale in biomedical and health research

Economies of Scope: Effect on research outputs of varying the scope of activities was less often reported in empirical studies than the effect of scale. *Taken as a whole*: The empirical literature more often suggests the existence of positive economies of scope, but the picture is mixed

•Largely focused at the whole-university or whole-institute level

Literature review results published in: Hernandez-Villafuerte, K., Sussex, J., Robin, E., Guthrie, S. and Wooding, S., 2017. *Economies of scale and scope in publicly funded biomedical and health research: evidence from the literature*. Health Research Policy and Systems. 15(3).

Acknowledgements: This research was supported with funding from the UK Medical Research Council (MRC). The funders played no role in the study design, data collection, analysis and interpretation, or writing of the manuscript.