Cost-effectiveness analysis of new diagnostic tools for cutaneous leishmaniasis in Afghanistan

Céline Aerts[1], Martijn Vink[2], Sayed Jalal Pashtoon[4], Sami Nahzat[5], Isra Cruz.[3], Elisa Sicuri[1,6]

[1]Barcelona Institute for Global Health, Barcelona, Spain;
[2]HealthNet TPO, Amsterdam, The Netherlands;
[3]Foundation for Innovative New Diagnostics, Geneva, Switzerland;
[4]HealthNet TPO, Kabul, Afghanistan;
[5]National Malaria and Leishmaniasis Control Program, Ministry of Public Health, Kabul, Afghanistan;
[6] Health Economics Group, Department of Infectious Disease Epidemiology, School of Public Health, Imperial College London, London, UK

ISGlobal Instituto de Salud Global Barcelona

EuroLeish.net Training Network

Ministry of Public Health Islamic Republic of Afghanistan

Introduction

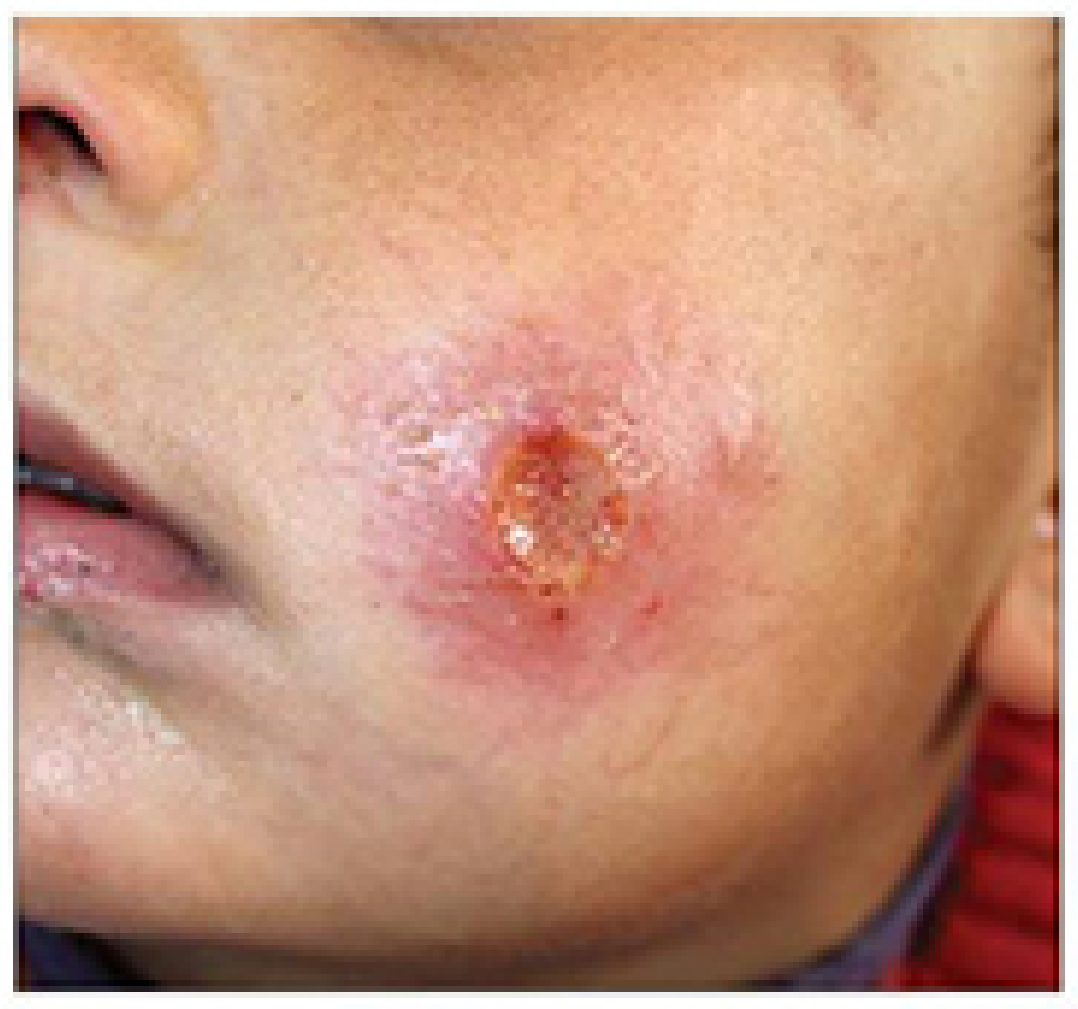

- Cutaneous Leishmaniasis (CL) is endemic in 98 countries but approximately 75% of the cases are concentrated in 10 countries
- Afghanistan is one of the countries with the highest burden
- The diseases produces lesions on exposed parts of the body (sometimes up to 200). It is responsible for chronic and disfiguring skin lesions resulting in morbidity and social stigma

What about the economics of CL?

In general, the literature on the economics of CL is very poor; the economic loss due to CL at the national and individual level is unclear. Similarly, the social burden inherent to this disease is not well quantified and has been largely underestimated.

Research question

Can any of these tools (RDT or LAMP) be a cost-effective alternative to microscopy?

An economic evaluation of these new diagnostic tools under different plausible scenarios (e.g. varying the sensitivity and specificity estimates, adherence to treatment, disability burden attributed to CL, etc.) can go some way towards achieving accurate and early diagnosis and treatment, which are essential to reduce the reservoir of infection and control CL, thereby limiting economic loss and social burden

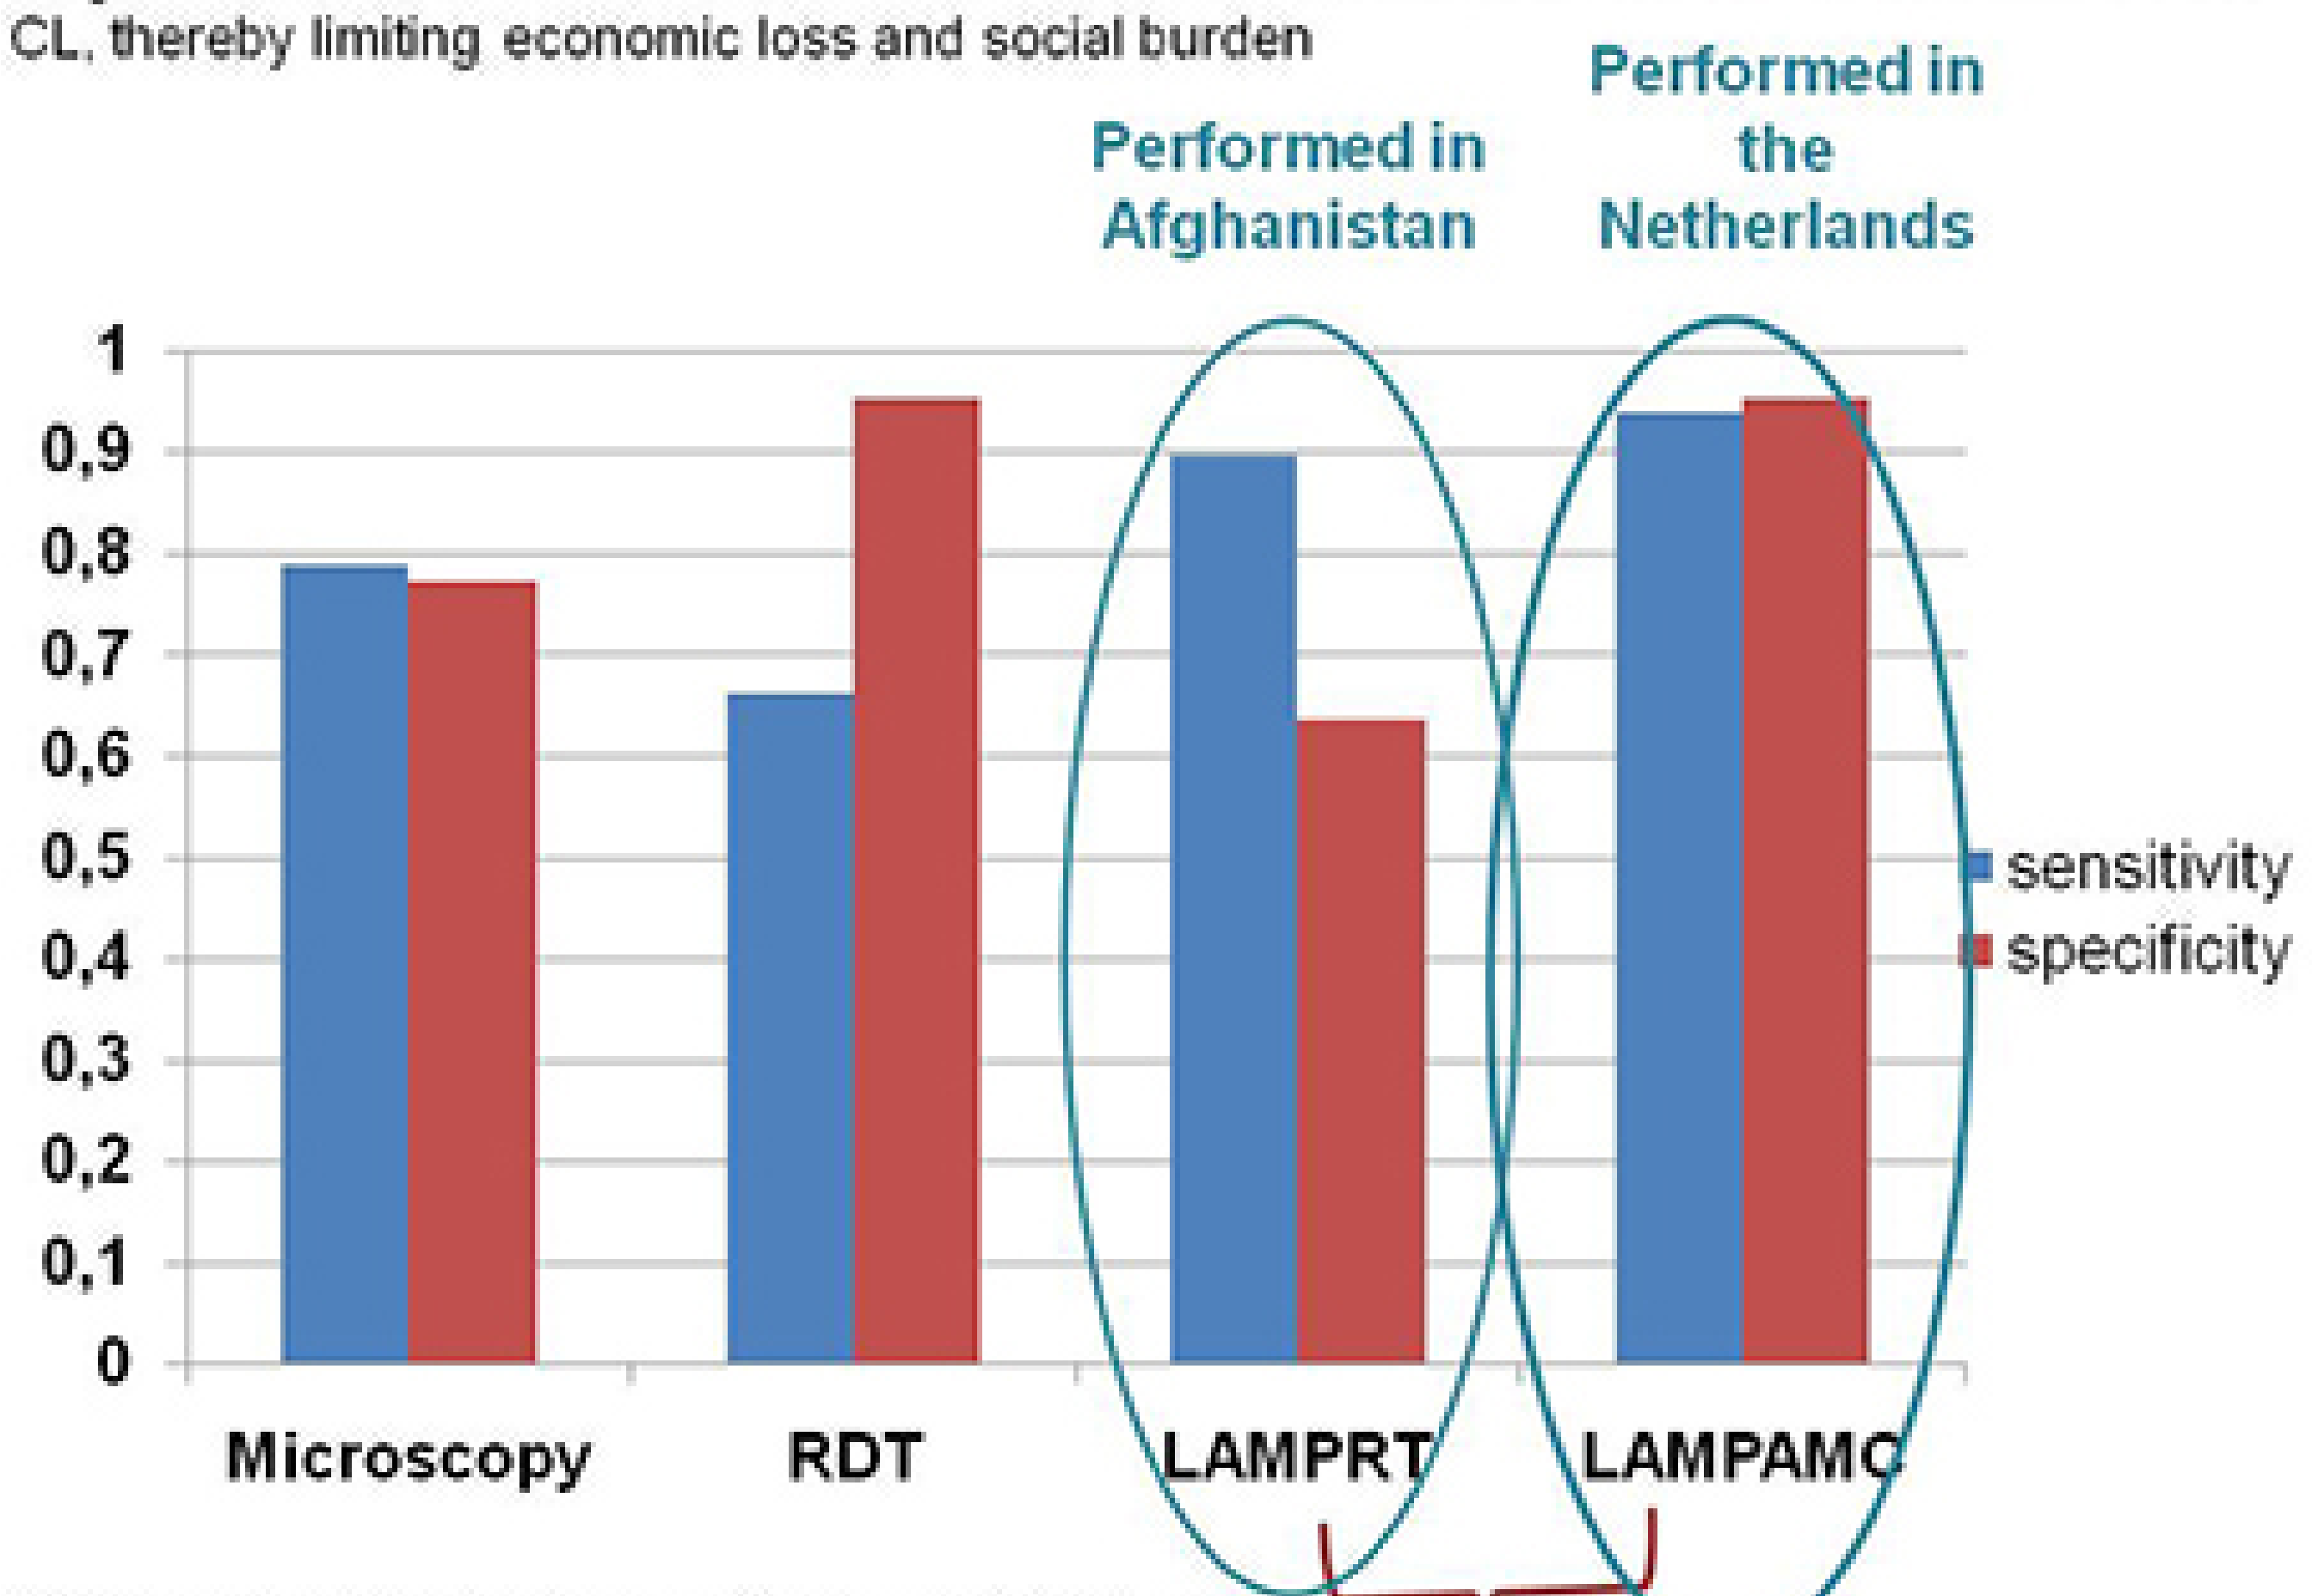
Performed in Afghanistan
Performed in the Netherlands
1
0,9
0,8
0,7
0,6
0,5
0,4
0,3
0,2
0,1
0
Microscopy
RDT
LAMPRT
LAMPAMC
sensitivity
specificity
Same tool

Source:National Malaria and Leishmaniasis Control Programme (NMLCP) and Health Protection and Research Organization (HPRO)

Context

The current diagnostic tools for CL are:

Microscopy

- ✗ Low and variable sensitivity
- ✗ Requires trained personnel

PCR

- ✗ Well equipped laboratory facilities and experienced personnel
- ✗ Costly

➡ There is a need for diagnostic tools with **high accuracy** that are **cheaper**, **easier to use** and **field-amenable** in order to reduce morbidity, relapses and risk of anthroponotic transmission.

Two new point-of-care diagnostic tools have been recently developed:

CL Detect Rapid Test (RDT)
(InBios International)

- ✓ A close to perfect sensitivity
- ✓ Easy and cheap to use
- ✗ variable sensitivity

Loopamp detection kit (LAMP)
(Eiken Chemical Co)

- ✓ Performing equally well as of PCR
- ✓ Simpler to use
- ✗ Can be costly to implement

Data and Model

Data were collected between March and July 2016 among Individuals presenting themselves at the health facility (*The National Malaria and Leishmaniasis Control program*) in Kabul, with suggestive signs of CL.

Cost estimates: were taken from 3 questionnaires:

1. *"Patient cost questionnaire"*
2. *"Laboratory and medical staff questionnaire"*
3. *"Drug and treatment questionnaire"*

Effectiveness estimates: sensitivity and specificity of the tools AND Disability Adjusted-Life Years (DALYs)

The natural history of the disease was represented through a **Markov model** (Annual cycles; cf graph below), which was combined with a bigger decision tree designed in TreeAge

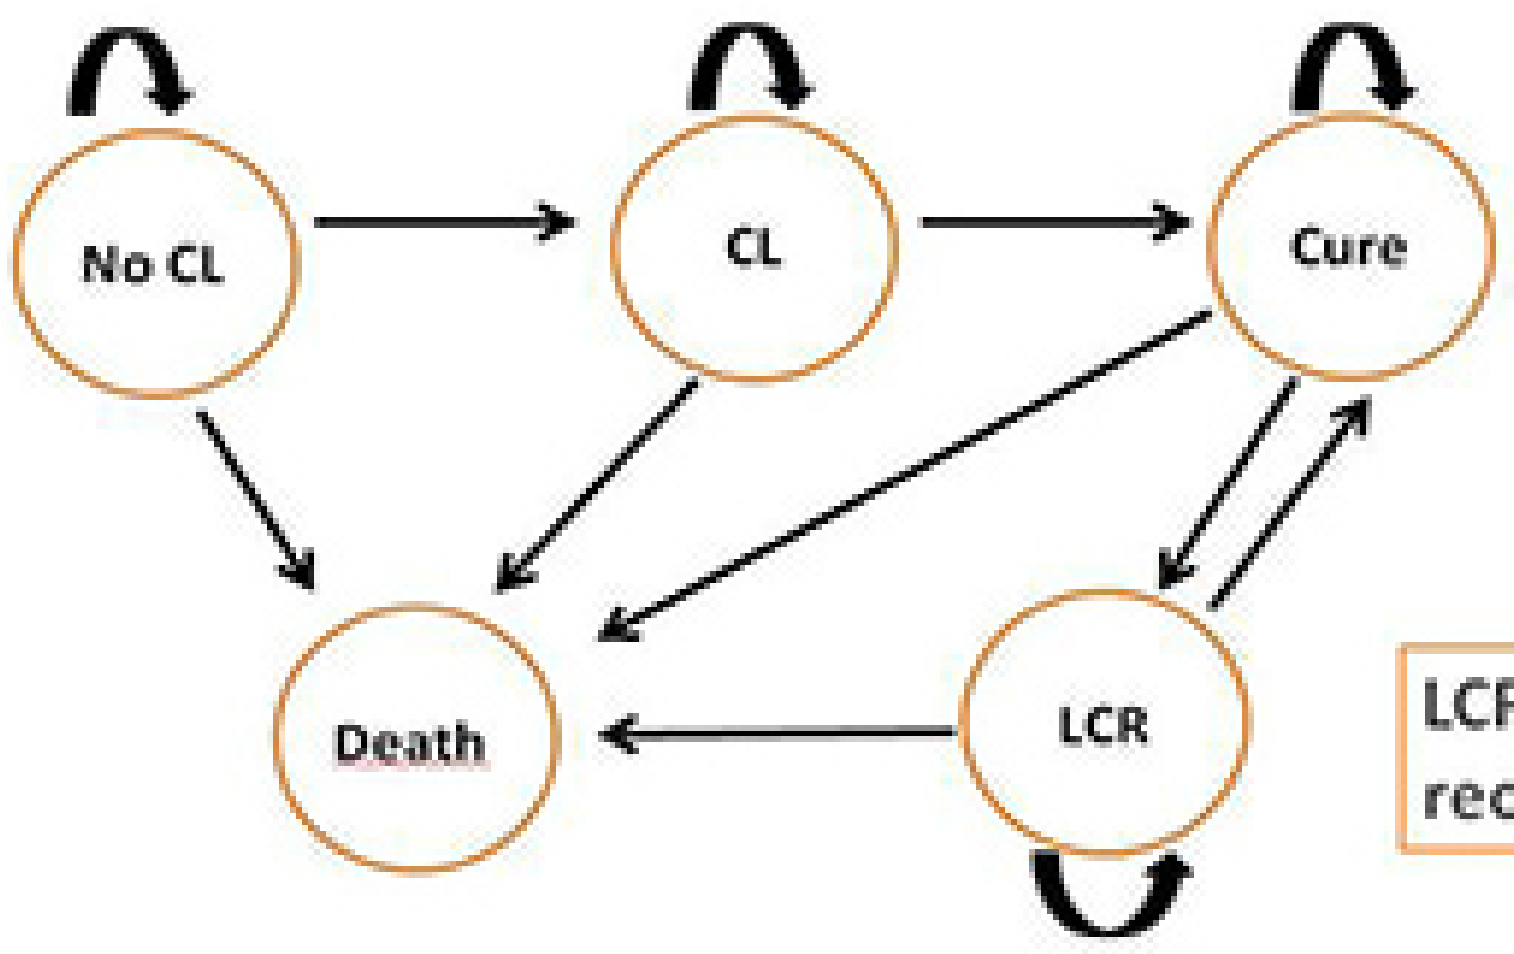
No CL
CL
Cure
Death
LCR

- ✓ Put a cohort of 10 000 individuals entering at age 5
- ✓ Run the model for 80 cycles

LCR= cutaneous leishmaniasis recidivans

Measurement of cost-effectiveness

How do we know if the new tool is cost–effective?

IF the incremental cost-effectiveness ratio (ICER) is < willingness-to-pay threshold

$$\frac{(\text{cost}_{\text{new strategy}} - \text{cost}_{\text{old strategy}})}{(\text{effectiveness}_{\text{new strategy}} - \text{effectiveness}_{\text{old strategy}})}$$

1 time the GDP per capita (**561 USD**)

3 times the GDP per capita (**1683 USD**)

Deterministic results (1)

For deterministic analyis we use the mean or the median value for each parameter

	Average per person		ICER	Outcome
	Cost (2016 USD)	DALY	USD per DALY averted	
Microscopy	67.19	3.63		undominated
LAMPAMC	74.93	3.60	**303.32** <WTP	undominated
RDT	67.41	3.65	-10,15	dominated
LAMPRT	75.09	3.61	-22,38	dominated

The ICER is < both WTP tresholds: LAMPAMC is a cost-effect. alternative to microscopy

Probabilistic results (2)

For probabilistic analyses, we assign different probabilistic distributions to parameters to account for uncertainty. Such analyses are helpful to test the robustness of the deterministic results (1).

Cost-effectiveness plane: LAMPAMC vs Microscopy

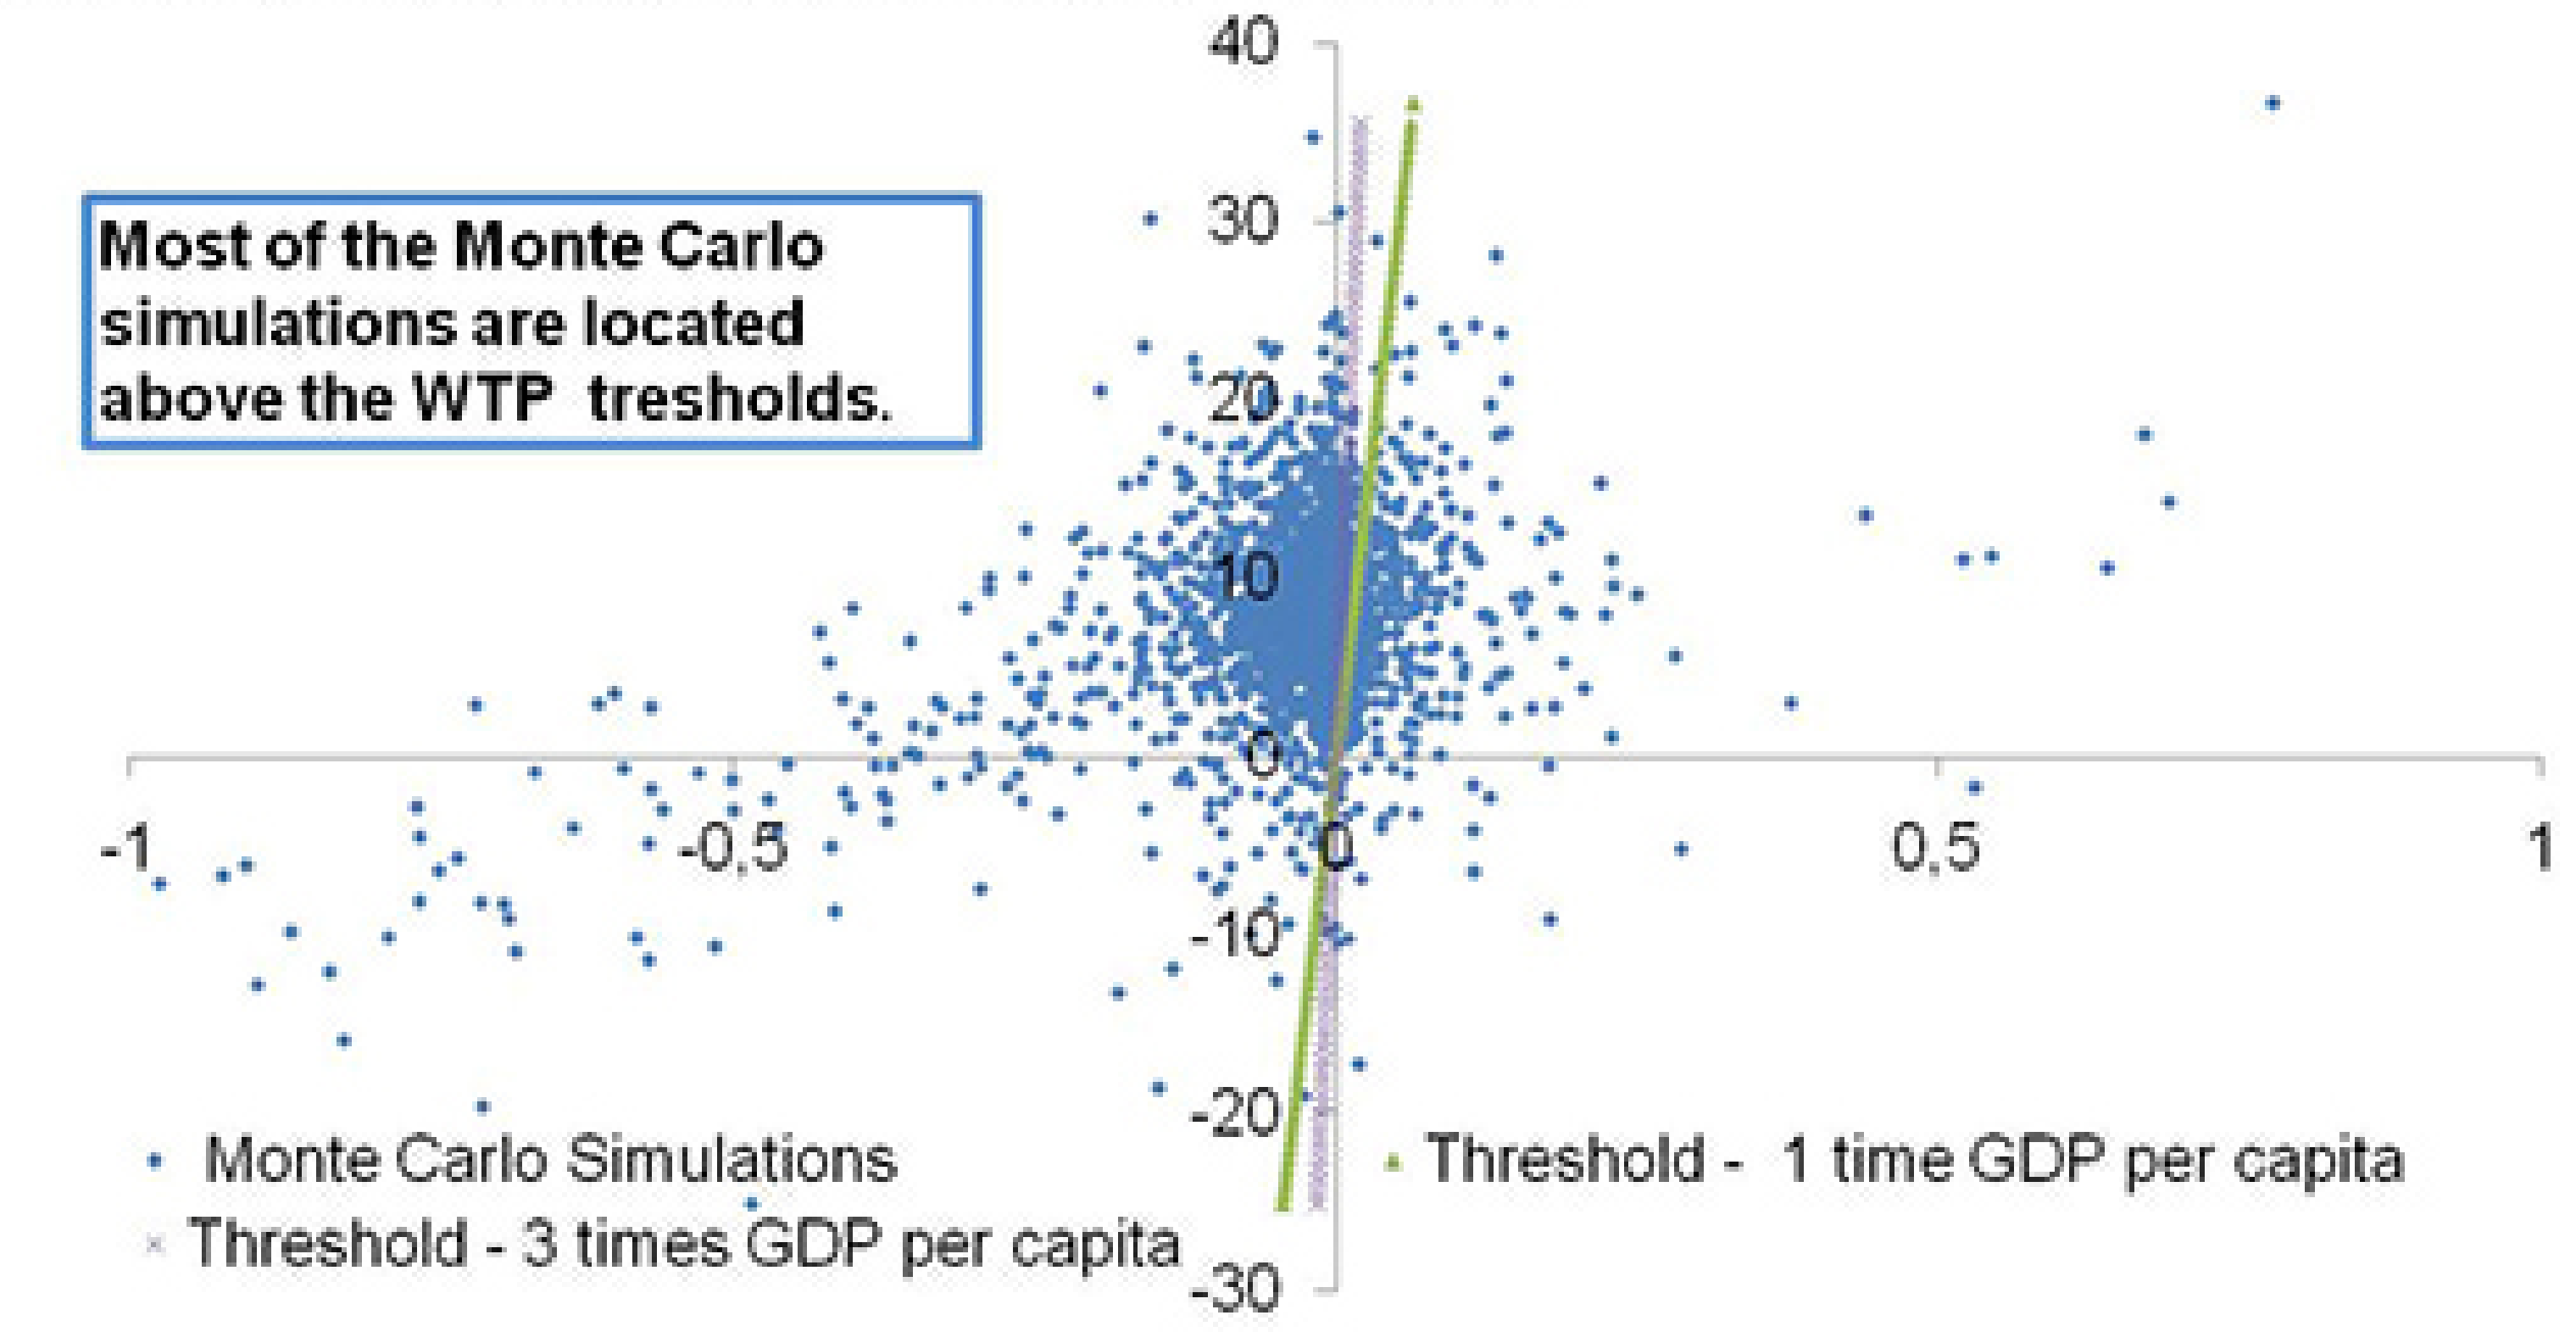
Most of the Monte Carlo simulations are located above the WTP tresholds.
40
30
20
10
0
-10
-20
-30
-1
-0,5
0
0,5
1
Monte Carlo Simulations
Threshold - 3 times GDP per capita
Threshold - 1 time GDP per capita

Uncertainty regarding the cost-effectiveness of LAMPAMC over microscopy is significant: probabilistic analyses favor the use of microscopy instead.

Conclusion

This study shows the potential of the LAMP to replace microscopy in the Afghan context. However this potential is weakened by the significant uncertainty surrounding its cost-effectiveness, as demonstrated by sensitivity analyses.

"This project has received funding from the European Union's Horizon 2020 research and innovation programme under the Marie Sklodowska-Curie grant agreement N° 642609"