AES 2017 póster nº 75

MASTERS PROGRAMS OF HEALTH ECONOMICS IN SPAIN: PROFILE OF THE GRADUATES AND JOB CONSEQUENCES

M Ruiz-Adame[a,b], M Correa[a,c], N García-Agua Soler[a,d]

a. AMES – Old Fellows Association of the Masters in Health Economics of the University of Málaga
b. Executive Function SL
c. Department of Applied Economics, Faculty of Economics Sciences and Business, University of Granada, Granada, Spain
d. Health Economics & Rational Use of Drugs, Faculty of Medicine, University of Málaga, Málaga

Human capital theory considers training and experience an investment. People with a higher level of education are given greater opportunities in the job market or better access to jobs.

Objective:
To identify the characteristics, motivations and working implications that appear among the graduates in Master Programs in Health Economics (MPHE).

Method:
The most renowned MPHE of Spain were contacted to request assistance in this research. Participants submitted an online survey comprised of 30 items designed specifically for the purpose. 1319 e-mails were sent in total. Our sample was formed of 438 graduates.

Results:
The main motivation for doing a MPHE is academic. People do it in order to acquire new or to enhance previous knowledge. The general profile of graduates is that of a woman of 37.8 years old who is a health professional. The people who looked for a job in Health Economics found one mainly in the first (54.9%) or second year (29.7%). The evaluations of the MPHEs are very highly regarded. The most useful subject was health management (46.3%).

Table 1: Masters programmes in health economics that participated in the research. Rate of answer/email sent

MPHE	Year beginning the MPHE	Num. of academic Programmes	Num. of Graduates/ Programme (Average)	Total Graduates	Num of Emails sent	Answers to the survey	Rate of answer/ email sent
AEPH	1984	32	31	1010	600	167	0.2783
UPF	2004	12	44	528	304	98	0.3224
UMA	2008	8	20	160	127	70	0.5512
UNICAN	2009	7	30	210	213	58	0.2676
UCL-MWI	2012	4	19	76	75	40	0.5333
Others						6	
TOTAL		63		1614	1319	438	0.3321

Source: Own elaboration from data.
MPHE: Masters Programmes in Health Economics; AEPH: Andalusian School of Public Health; UPF: University Pompeu Fabra; UMA : University of Málaga; UNICAN: University of Cantabria; UCL : University of Castilla-La Mancha; MWI: Max Webber Institute

Table 2: Working situation and expectation of graduates. General group vs graduates who found a job in health economics after the MPHE.

Variable/ Question	Values	Most common answer: General sample (N=438)	Most common answer: Graduates who found a job in HE after the MPHE (n=91)
Working situation at the moment of enrolment in the MPHE	1: Unemployed. Searching for a job 2: Unemployed, but not looking for a job 3: Employed 4: Employed, looking to improve their education 5: Employed, looking to change their job 6: Employed, looking for a promotion from their job 7: Recipient of scholarship	3: Employed (37.9%) 4: Employed, looking to improve their education (33.3%)	4: Employed, looking to improve their education (35.2%) 3: Employed (25.3%)
Working situation at the moment of enrolment in the MPHE Classified by : Unemployed, Employed, Recipient of scholarship	0: Unemployed 1: Employed 2: Recipient of scholarship	1: Employed (87.0%)	1: Employed (75.8%)
Did you consider that the MPHE would help you to improve your working situation? (to get promoted in your job, or to get a job…)	0: No 1: Yes	1: Yes (85.4%)	1: Yes (93.4%)
Have you ever had employment related to HE after the MPHE?	0: No 1: Yes	1: Yes (53.0%)	1: Yes (100.0%)
Did the MPHE help you to get a job in the field of HE?	1: No. I have never looked for a job in this field 2: No. I have not got a job in this field 3: No. I was already working in this field. 4: Yes	3: No. I was already working in this field. (33.1%)	4: Yes (100.0%)
Did the MPHE help you to get a job in the field of HE? Classified by : Yes or No	0: No 1: Yes	0: No (79.2%)	1: Yes (100.0%)

Table 3: Characteristics of the job found by the graduates in the field of HE (for those who found a job after the MPHE)

Variable/ Question	Values	Most common answer: Graduates who found a job in HE after the MPHE (n=91)
Years between the end of the MPHE and the graduate getting a job	Years 1 If it was found in the same year. 2 if it was found in the next year	1 (54.9%)
Period of contract	1: Between 1 and 3 months 2: Between more than 3 and 6 months 3: Between more than 6 and 12 months 4: More than 12 months 5: Permanent 6: For a specific project 7: Freelance	5: Permanent (37.4%)
Net revenues (per month)	1: Less than 500€ 2: Between 500€ and 1000€ 3: Between more than 1000€ and 1,500€ 4: Between more than 1,500€ and 2,000€ 5: Between more than 2,000€ and 2,500€ 6: More than 2,500 €	4: Between more than 1,500€ and 2,000€ (27.5%)
What do you consider as the most useful in getting the job?	1: Personal contacts 2: Contacts with other fellows 3: Contacts with professors 4: The training that I received, my CV 5: Other	4: The training that I received, my CV (71.4%)

Table 4: Evaluation of the MPHE by the graduates. General group vs graduates who found a job in HE after the MPHE.

Variable/ Question	Values	Most common answer: General sample (N=438)	Most common answer: Graduates who found a job in HE after the MPHE (n=91)
Evaluation of the training on the whole	1 to 10	8 (29.7%)	8 (28.6%)
Evaluation of the multidisciplinary training	1 to 10	8 (26.9%)	9 (30.8%)
Degree of applicability to real life of the knowledge obtained	1 to 10	8 (25.3%)	8 (27.5%)
What kind of knowledge area do you think was most useful ?	1: Data analysis 2: Epidemiology 3: Research methodology 4: Economics 5: Health management 6: Law 7: Health 8: Social 9: Other	5: Health management (46.3%)	5: Health management (40.7%)
Evaluation of the training in order to increase employability	1 to 10	8 (21.0%)	8 (27.5%)
Do you keep in contact for labour reason with old fellows?	0: No 1: Yes	1: Yes (55.3%)	1: Yes (57.1%)
Type of financing of the training	1: Partially funded by a private company 2: Totally funded by a private company 3: Partially funded by the public sector 4: Totally funded by the public sector 5: Totally personally funded	5: Totally personally funded (36.3%)	5: Totally personally funded (37.4%)

(*) Lower punctuation means lower satisfaction.

Conclusions:
To do a MPHE is a good investment because most of the graduates believe that they got a job thanks to their training. Graduates show a high degree of confidence in the utility of training. The MPHE are highly evaluated independent of the job consequences. The subjects in which the curriculum vitaes of the health professionals are weaker, such as those concerning management, are the best valued as they are assumed to enhance promotion opportunities.